Engineering a Bispecific Claudin 18.2/TGF-β CAR to Target Claudin 18.2 Tumors and Overcome the Tumor Microenvironment

Jessica Reyes, Ethan BenDavid, Orit Foord, Ximin Chen, Michael Weist, Giulia Parisi, Jiajia Cui, and Melanie L. Munguia

ImmPACT Bio, West Hills, California, United States of America. Contact: info@immpact-bio.com; (818) 857-4828

Poster Number:832

Abstract

Gastric cancer (GC) is among the top three deadliest cancers and has the fifth-highest global cancer incidence (1). Claudin 18.2 (CLDN 18.2) is an isoform of the tight-junction protein Claudin 18 and is emerging as a promising target for CAR T cell therapies against GC. CLDN18.2 shows limited expression in normal tissues but is upregulated during malignant transformation as seen in GC, making CLDN18.2 an attractive tumor-associated antigen. (2). Recently, a clinical trial (NCT03874897) used CLDN18.2 CAR T cells to treat patients with GC and pancreatic cancer. In the GC population, the overall response rate and disease-control rate were 57.1% and 75.0%, respectively, with a median progression-free survival of 4.2 months among 28 treated patients (3). Repeat infusions of the CAR T product was provided to 39% of patients, which speaks to the potential need to enhance T cell persistence and function in solid tumors.

Studies have shown that GC patients have elevated levels of transforming growth factor-beta (TGF-β) in their gastric mucosa (4). This highly pleiotropic cytokine has profound immunosuppressive effects that limit T-cell persistence while also promoting tumor survival, immune evasion, and metastasis. Moreover, TGF-β induces the differentiation of CD4+ T cells into regulatory T cells (Tregs) that in turn attenuate immune activation and production of proinflammatory mediators (5). We hypothesized that, by engineering T cells to convert the TGF-β anti-inflammatory signal into a pro-inflammatory signal, we could significantly ameliorate TGF-β's inhibitory effects and enhance T-cell function, proliferation, and infiltration of CAR T cells in gastric tumors.

We designed a bispecific, OR-gate CAR to target both CLDN18.2 and TGF-β. The CLDN18.2/TGF-β bispecific CAR T cells were co-cultured with either CLDN18.2-positive gastric-cancer cell lines or with different isoforms of TGF-β. When co-cultured with CLDN18.2 cell lines, both the single-input (CLDN18.2 only) and bispecific CAR T cells showed high cytotoxicity with similar levels of T-cell activation, as evaluated by IFN-γ secretion. In addition, bispecific CAR T cells demonstrated reduced exhaustion markers and fewer Tregs compared to the CAR T cells that solely target CLDN18.2.

In summary, we show that the CLDN18.2/TGF-β bispecific CAR converts the immunosuppressive TGF-β signal into an activation signal, thereby enhancing T-cell fitness, reducing T-cell conversion into Tregs, and decreasing phenotypic exhaustion.

Methodology

Healthy-donor leukapheresis products were processed to enrich CD62L+ T cells. These enriched cells were transduced using a lentiviral vector system, which encoded a bispecific CAR targeting CLDN18.2 and TGF-β (IMPT-601). Control CAR T cells were simultaneously produced to express either a CLDN18.2 single-input CAR[1] or a benchmark CAR[2]. Following the manufacturing process, we harvested the CAR+ T cells and evaluated specificity, cytotoxicity, and phenotypic fitness.

CAR T cells were co-cultured with either recombinant TGF-β protein or an engineered human gastric cancer cell line (AGS) that expresses either of the CLDN18 isoforms (CLDN18.1 or CLDN18.2). Effectiveness of the CAR construct was assessed by measuring T-cell proliferation and IFN-γ secretion.

To assess T-cell fitness, T cells were co-cultured with a human GC cell line (GSU) that endogenously expresses CLDN18.2 and secretes active TGF-β. Two in vitro systems were utilized: repeated antigen challenges (RACs) and a three-dimensional (3D) spheroid model. In the RAC system, co-cultures were established at a 1:1 ratio of CAR+ T cell : GSU. After 2-3 days of incubation, the T-cell–rich supernatants were harvested and transferred to a new GSU challenge. At each RAC, a 12-point effector-to-target (E:T) titration was concurrently established to monitor cytotoxic sensitivity.

Spheroids were generated by seeding GSU cells in a 96-well, ultra-low attachment (ULA), round-bottom plate containing basement membrane extract (BME) for 1-2 days. After spheroid formation, T cells were titrated and seeded, with the highest E:T ratio at 10:1.

T cells were evaluated for Treg differentiation and T-cell exhaustion at the endpoint of RAC experiments and spheroid co-cultures.

[1]CLDN18.2 single-input CAR: CLDN18.2 targeting CAR, containing the parental CLDN18.2-binding scFv of IMPT-601.

[2]Benchmark CAR: CAR containing a clinically evaluated CLDN18.2-binding domain.

Abbreviations: BME = basement membrane extract, GFP = green fluorescent protein, Hz = humanized, ULA = ultra low attachment, UTD = untransduced v/v = volume/volume

Schematic of IMPT-601

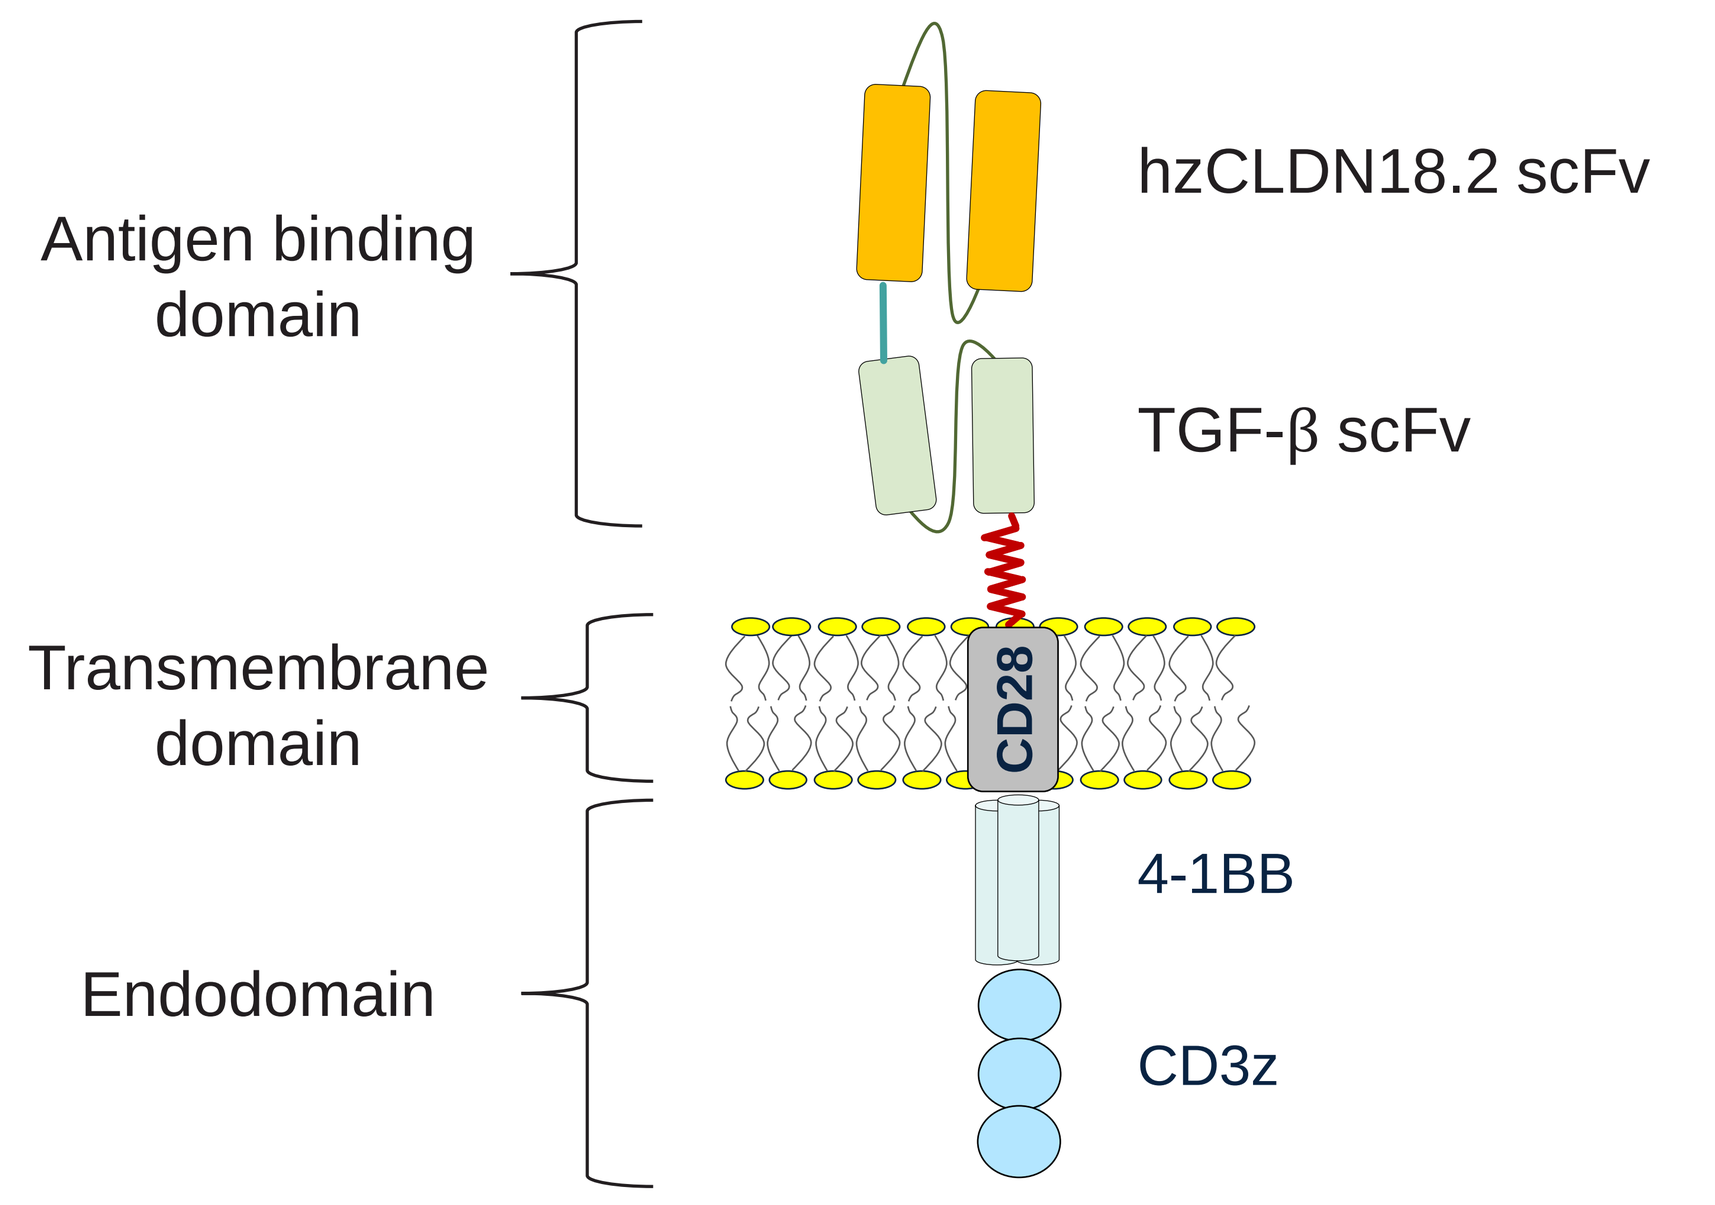
Antigen binding domain
hzCLDN18.2 scFv
TGF-β scFv
Transmembrane domain
CD28
Endodomain
4-1BB
CD3z

Figure 1. IMPT-601 is a second-generation, bispecific, OR-gate CAR. The extracellular domain is comprised of two linked single-chain variable fragments (scFvs) targeting CLDN18.2 and soluble TGF-β. IMPT-601 has a CD28 transmembrane domain, a 4-1BB co-stimulatory domain, and a CD3ζ activation domain.

IMPT-601 delivers target-specific T-cell activation in the presence of CLDN18.2 or TGF-β

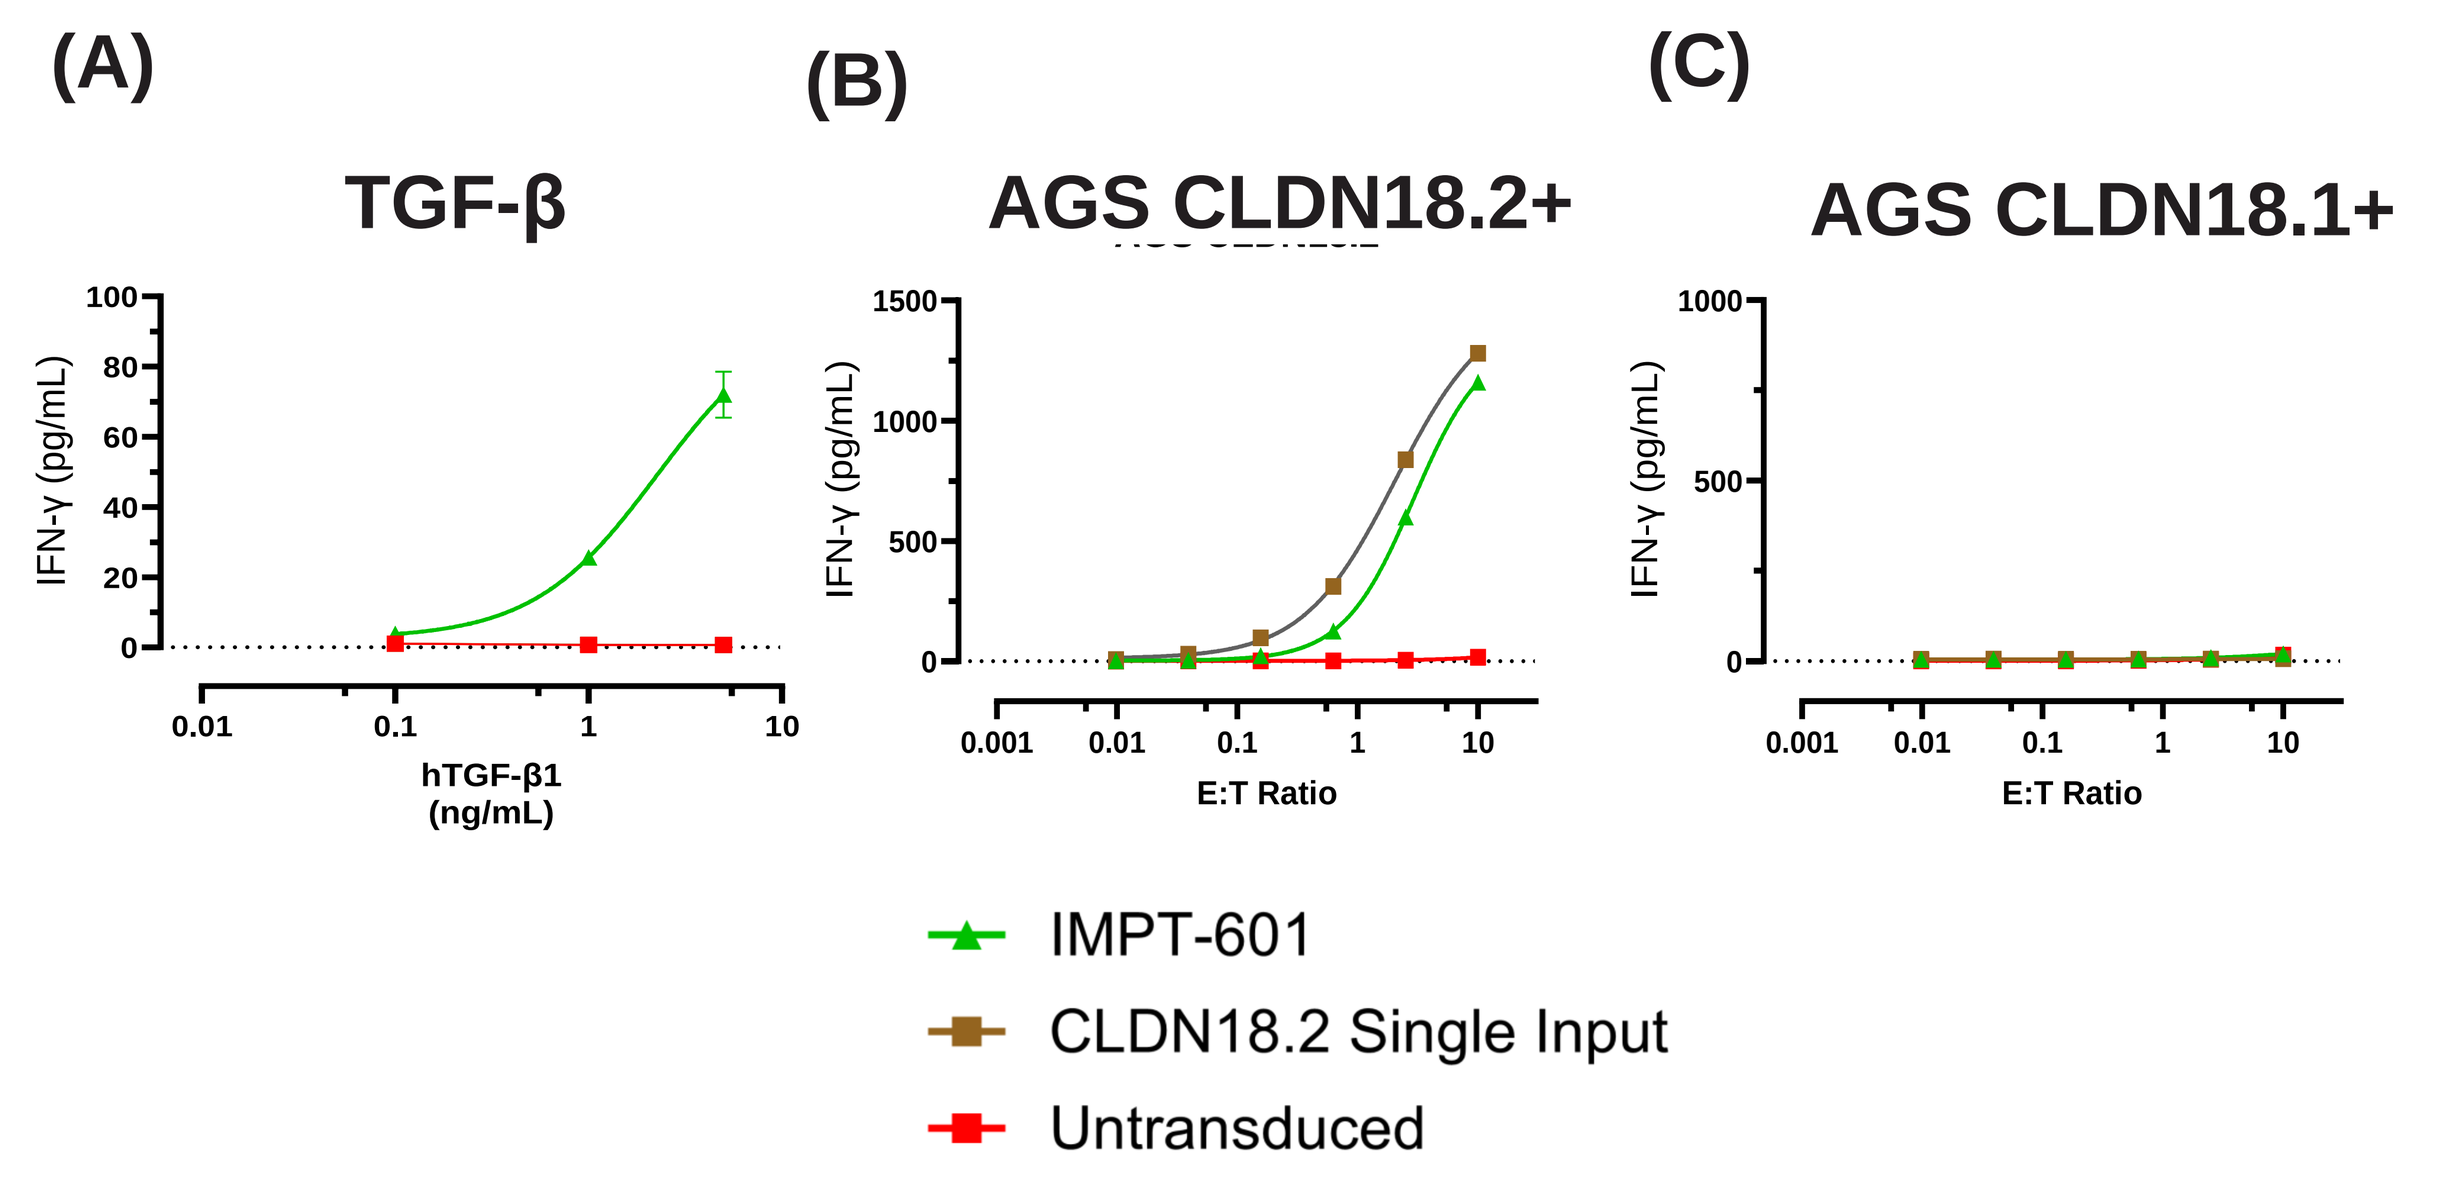
(A)
TGF-β
IFN-γ (pg/mL)
hTGF-β1 (ng/mL)
(B)
AGS CLDN18.2+
E:T Ratio
(C)
AGS CLDN18.1+
IMPT-601
CLDN18.2 Single Input
Untransduced

Figure 2. IMPT-601 CAR T cells demonstrate specific activation when cultured with either TGF-β or CLDN18.2. **(A)** T cells were spiked with varying concentrations of huTGF-β1. IMPT-601 CAR T cells secrete IFN-γ in a dose-dependent manner. **(B)** IFN-γ secretion of IMPT-601 CAR T cells co-cultured with AGS CLDN18.2+ gastric cell line. IMPT-601 and single input CLDN18.2 CAR T cells show similar levels of IFN-γ production. **(C)** Low/no detectable levels of IFN-γ levels were produced when co-cultured with the AGS CLDN18.1+ gastric cell line.

IMPT-601 demonstrates persistent cytotoxicity across repeated antigen challenges (RACs)

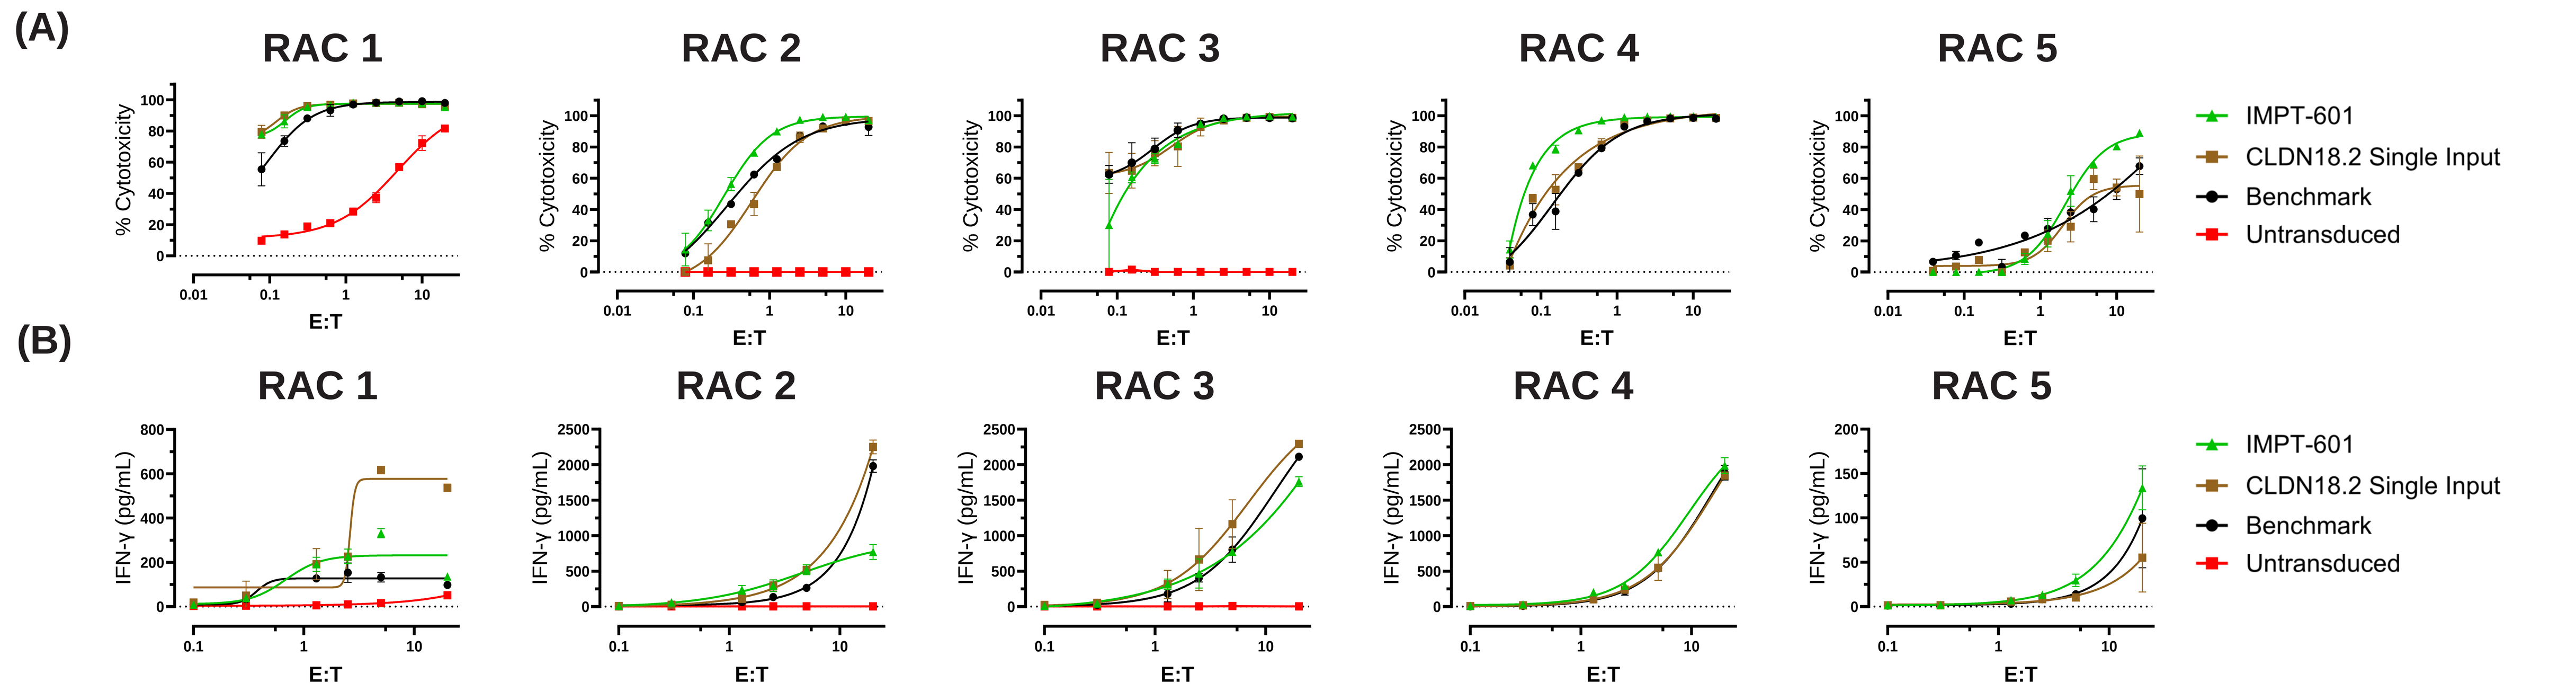
(A)
RAC 1
RAC 2
RAC 3
RAC 4
RAC 5
% Cytotoxicity
E:T
(B)
IFN-γ (pg/mL)
IMPT-601
CLDN18.2 Single Input
Benchmark
Untransduced

Figure 3. IMPT-601 is highly efficacious across five GSU RACs. **(A)** To establish T-cell killing potency, T cells were co-cultured with GSU gastric cell line across a 12-point E:T titration. At RAC 5, IMPT-601 CAR T cells demonstrate enhanced cytolytic characteristics compared to the other constructs, reaching nearly complete tumor clearance at higher E:T ratios. Single input control CARs, show a slight decrease in killing efficacy, reaching approximately 60-80% tumor clearance at the highest E:T ratio. **(B)** CAR+ T cells secrete IFN-γ at detectable levels when co-cultured with GSU gastric cell line. IFN-γ levels vary between challenges but are similar across most of the CAR T cell constructs. UTD T cells were discontinued at RAC 3 due to lack of T cell expansion.

IMPT-601 reduces regulatory T cell differentiation and phenotypic exhaustion

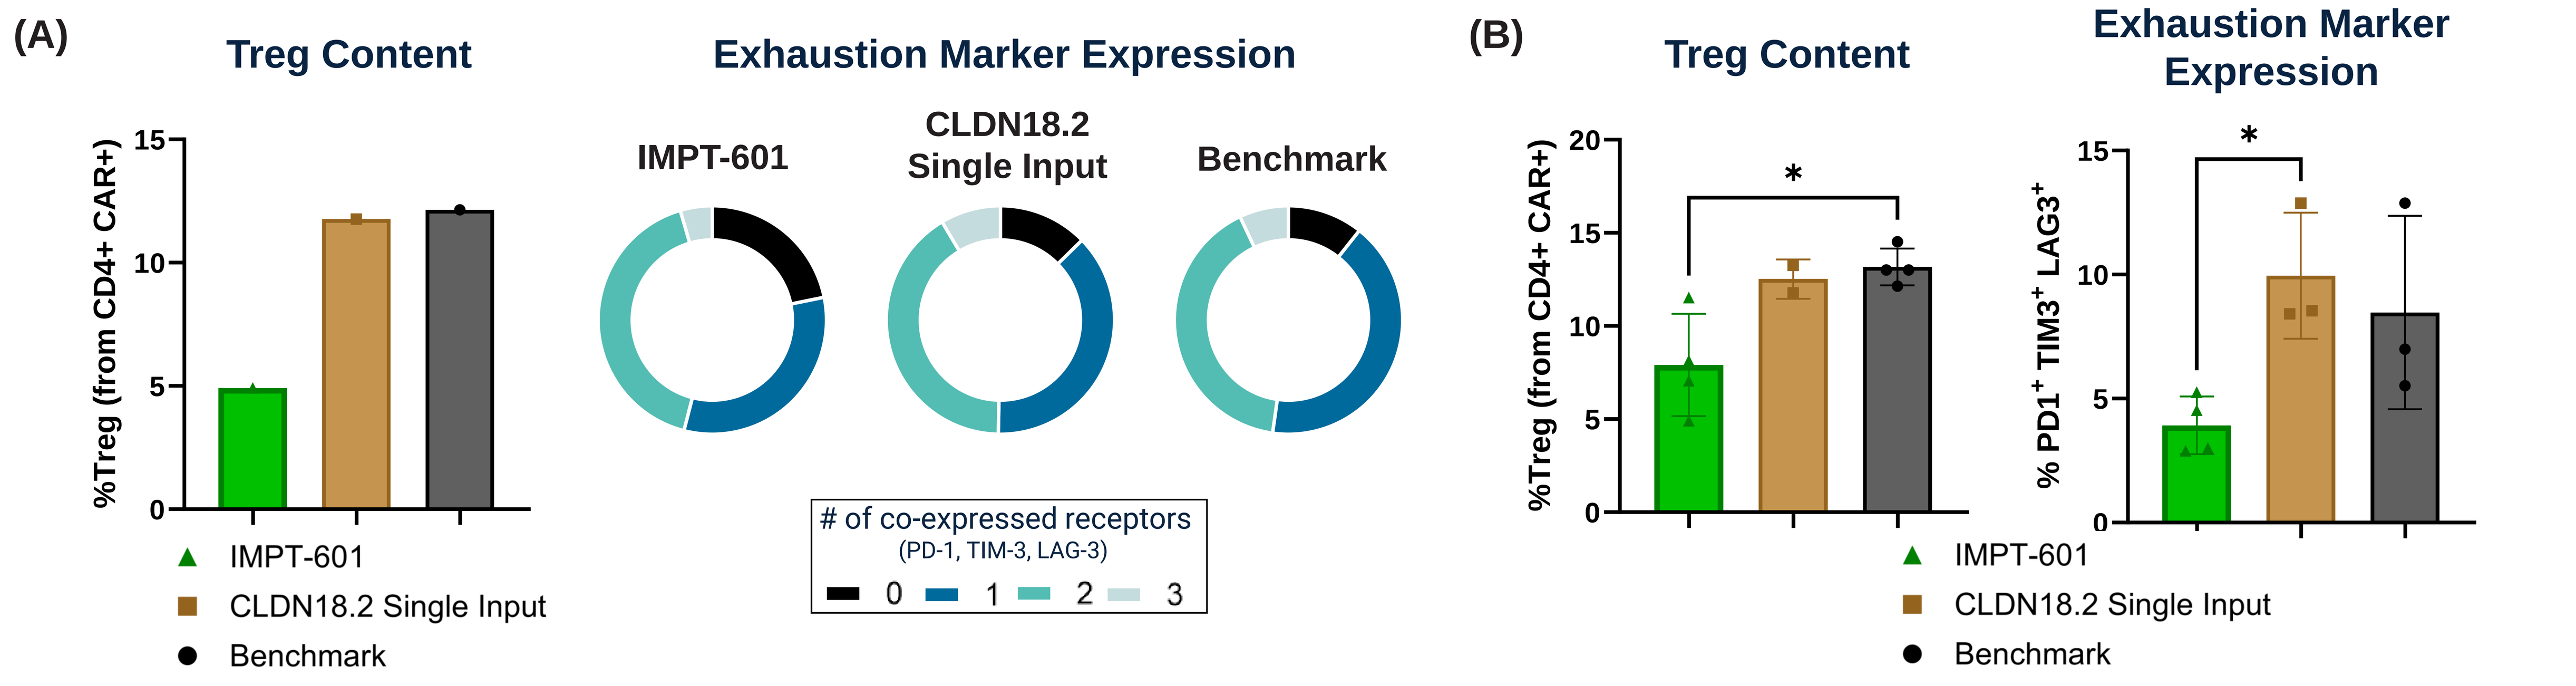
(A)
Treg Content
%Treg (from CD4+ CAR+)
Exhaustion Marker Expression
IMPT-601
CLDN18.2 Single Input
Benchmark
of co-expressed receptors
(PD-1, TIM-3, LAG-3)
0 1 2 3
(B)
Treg Content
Exhaustion Marker Expression
% PD1+ TIM3+ LAG3+
*
IMPT-601
CLDN18.2 Single Input
Benchmark

Figure 4. Regulatory and exhausted T-cell phenotypes were evaluated at the endpoint of all RACs using flow cytometry. **(A)** Representative Treg (left) and exhausted (right) T-cell quantification. **(B)** Treg content (left) and exhaustion marker expression (right) across three donors at the end of RAC5. In both (A) and (B), the IMPT-601 CAR T cells exhibit significantly reduced Treg content when compared to CLDN18.2 benchmark. Likewise, the co-expression of exhaustion markers in IMPT-601 CAR T cells is lower when compared to the CLDN18.2 single-input control. Means of triplicates ±1 standard deviation are shown. Statistical significance was calculated using unpaired one-way ANOVA. Tregs are defined as CD4+, CD25+, CD127- and FOXP3+

IMPT-601 completely clears tumor spheroids and exhibits marked reduction in exhausted and immunosuppressive T-cell phenotypes

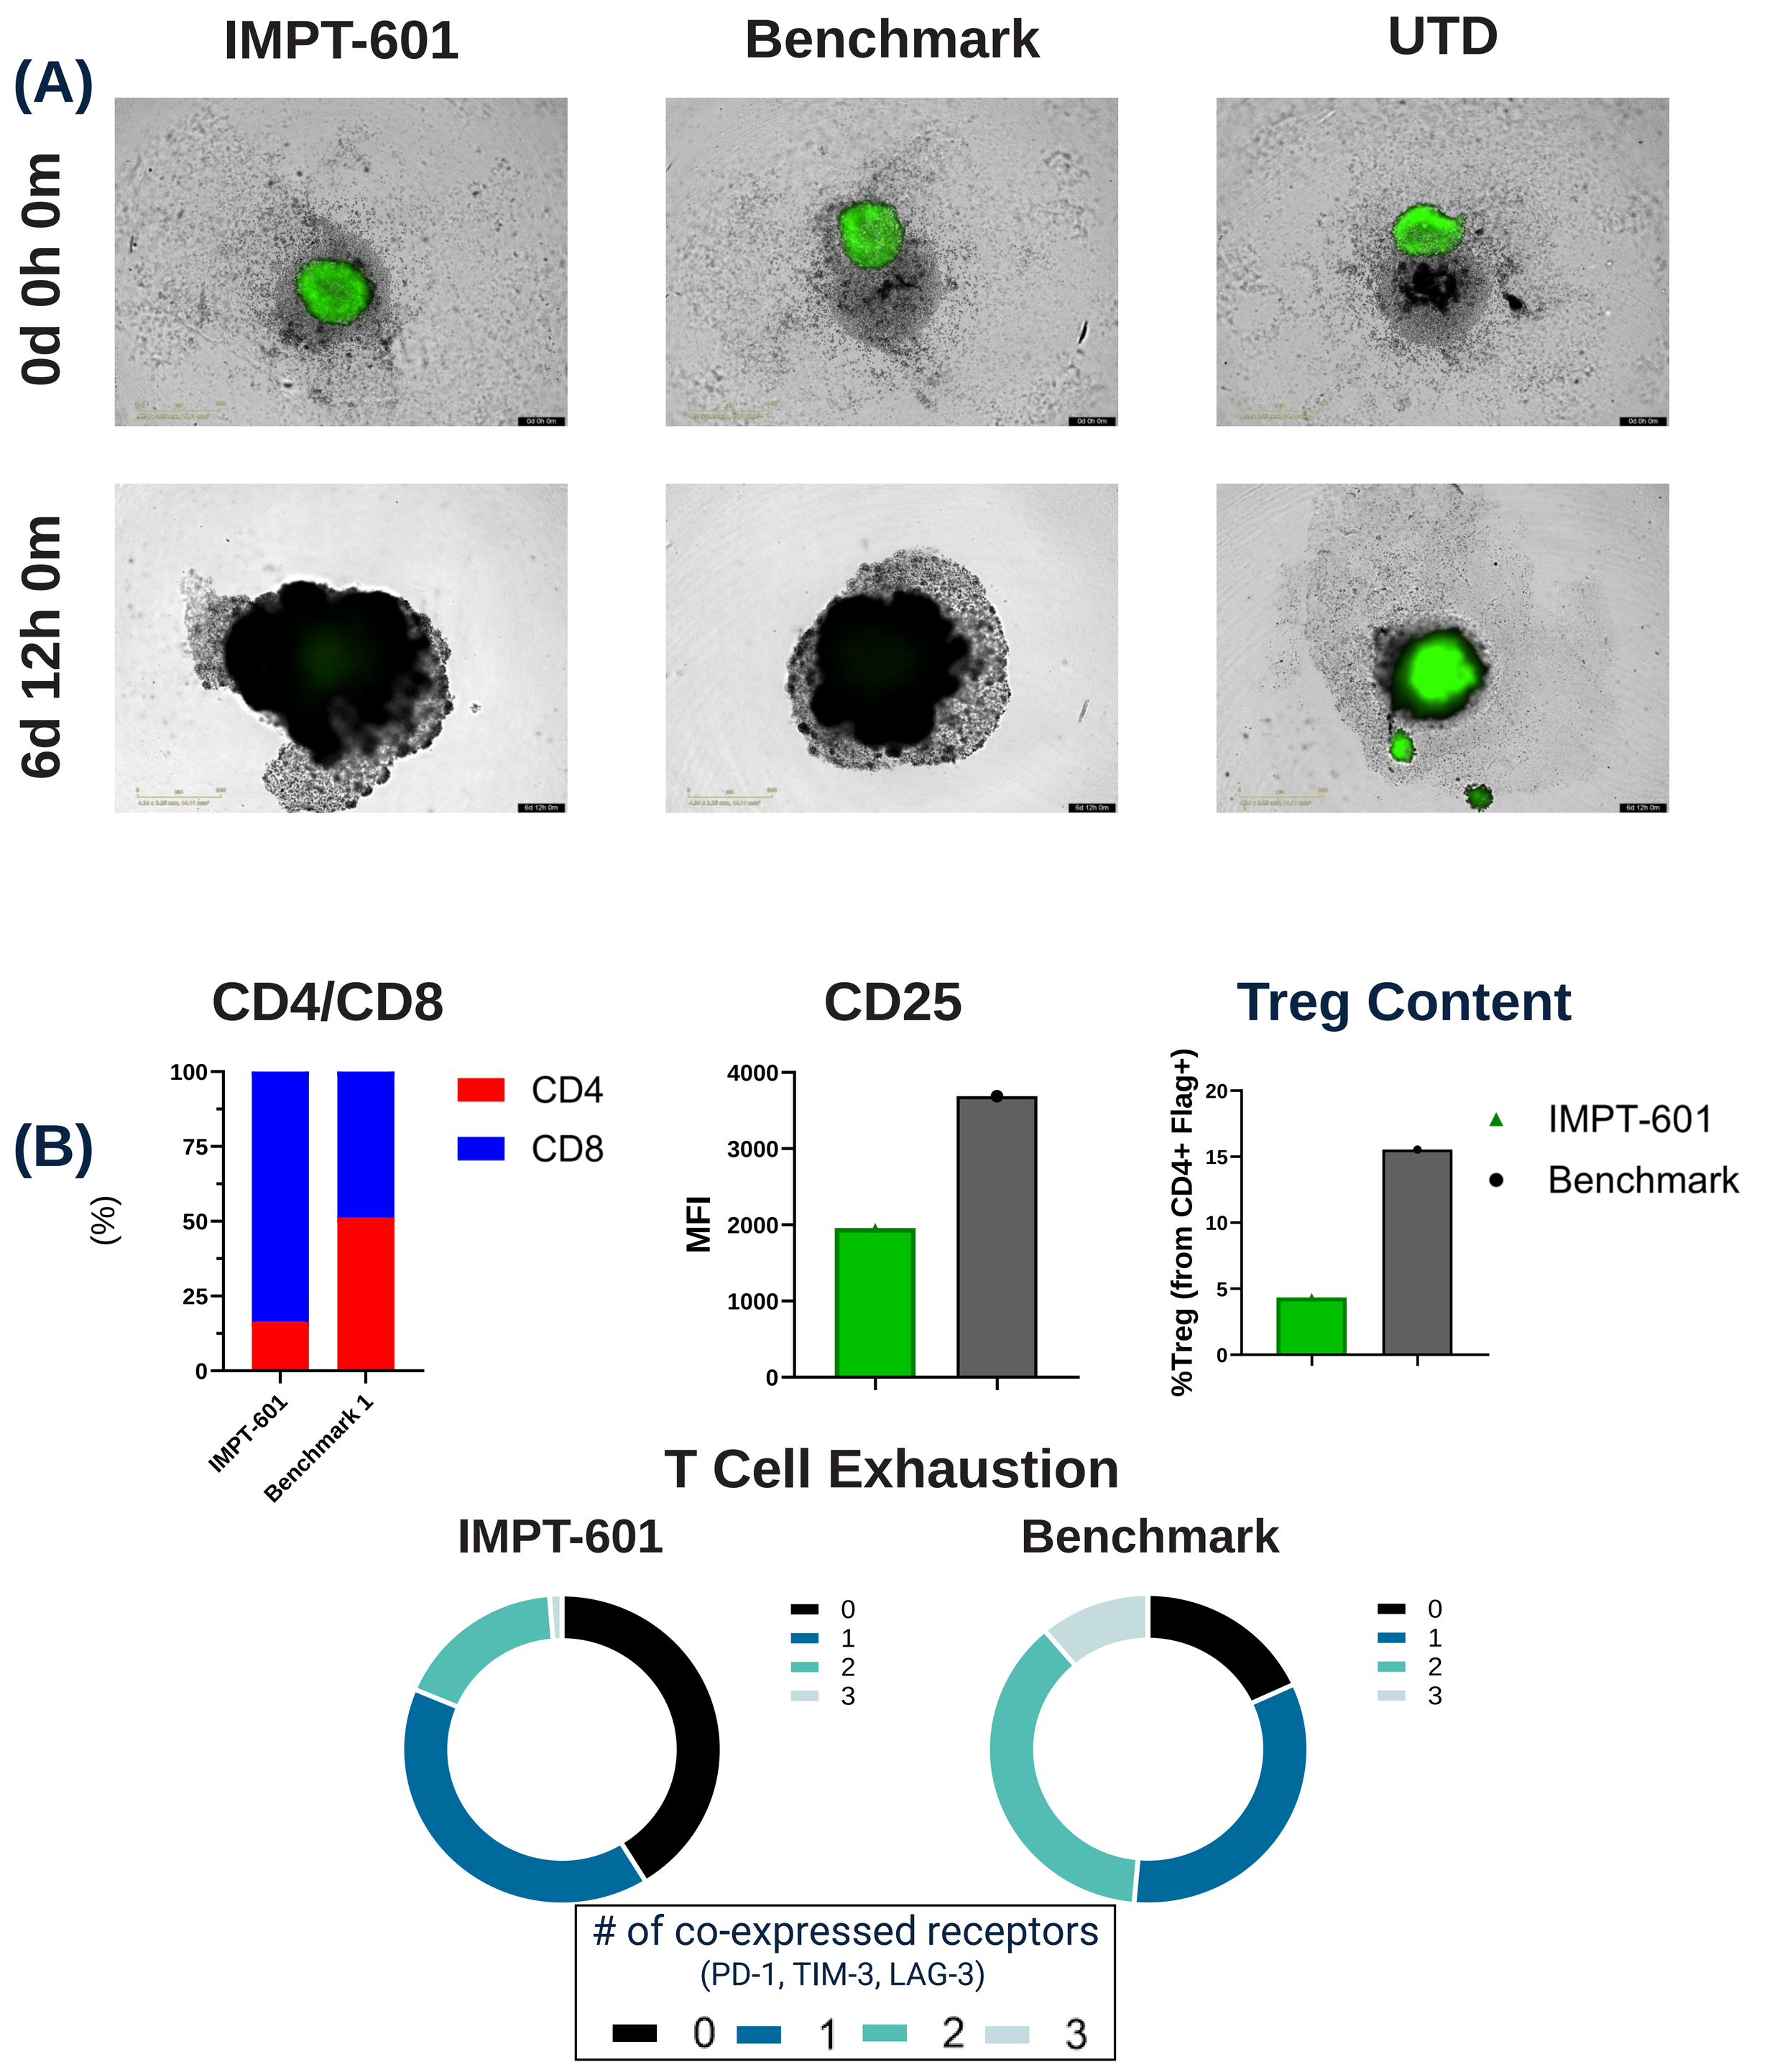
(A)
IMPT-601
Benchmark
UTD
0d 0h 0m
6d 12h 0m
(B)
CD4/CD8
CD4
CD8
CD25
MFI
Treg Content
%Treg (from CD4+ Flag+)
IMPT-601
Benchmark
T Cell Exhaustion
of co-expressed receptors
(PD-1, TIM-3, LAG-3)
0 1 2 3

Figure 5. Spheroids were generated by seeding GSU gastric cell line, expressing GFP, in a ULA 96-well plate at 3,000 cells/well with 2.5%(v/v) BME for 1-2 days. T cells were added to the GSU spheroids. **(A)** GSU cells (GFP) are cleared by CAR T cells after 6 days. Images were obtained by the Incucyte SX5. **(B)** IMPT-601 CAR T cells have lower %CD4, reduced CD25 MFI, and lower Treg content and exhaustion marker expression at experimental endpoint.

Conclusions

IMPT-601, a bispecific OR-gate CAR targeting both CLDN18.2 and TGF-β protein, demonstrated robust activation, proliferation, and IFN-γ secretion in response to either TGF-β protein or CLDN18.2 antigen. IMPT-601 reprograms an anti-inflammatory TGF-β signal into a pro-inflammatory signal, thus significantly enhancing anti-tumor immune responses in the immunosuppressive tumor microenvironment. Moreover, IMPT-601 maintained potent and sustained cytotoxicity against tumor cells over multiple rounds of antigen challenges. After 5 rounds of antigen challenges, IMPT-601 demonstrated reduced Treg differentiation and decreased exhaustion marker expression, suggesting that targeting of TGF-β protein, when combined with CLDN18.2 antigen targeting, could support persistent CAR T cell function despite extended antigen exposure. In summary, our data demonstrate the therapeutic potential of IMPT-601 against CLDN18.2-positive malignancies while overcoming the immunosuppressive tumor microenvironment.

References

1. Rawla, Prashanth, and Adam Barsouk. "Epidemiology of Gastric Cancer: Global Trends, Risk Factors and Prevention." Gastroenterology Review, vol. 14, no. 1, 2019, pp. 26–38, https://doi.org/10.5114/pg.2018.80001.
2. Hong, Jung Yong et al. "Claudin 18.2 expression in various tumor types and its role as a potential target in advanced gastric cancer." *Translational cancer research* vol. 9,5 (2020): 3367-3374. doi:10.21037/tcr-19-1876
3. Qi, Changsong et al. "Claudin18.2-specific CAR T cells in gastrointestinal cancers: phase 1 trial interim results." *Nature medicine* vol. 28,6 (2022): 1189-1198. doi:10.1038/s41591-022-01800-8
4. Luo, Jingwen, et al. "The role of TGF-β and its receptors in gastrointestinal cancers." Translational Oncology, vol. 12, no. 3, Mar. 2019, pp. 475–484, https://doi.org/10.1016/j.tranon.2018.11.010.
5. Hartley, Jordan, and Hinrich Abken. "Chimeric antigen receptors designed to overcome transforming growth factor-β-mediated repression in the adoptive T-cell therapy of solid tumors." *Clinical & translational immunology* vol. 8,6 e1064. 22 Jun. 2019, doi:10.1002/cti2.1064